Components of belief updating and their neural correlates

Nikki Marinsek, Benjamin Turner & Michael Miller
Psychological & Brain Sciences, University of California, Santa Barbara

Introduction

When expectations are violated, individuals revise or reject their current beliefs and search for new explanations[1,2,3]

We aimed to 1) identify brain regions that are sensitive to expectation violations and 2) build a Bayesian model of subjects' behavior in order to better understand the neural basis of hypothesis making.

fMRI procedure

Fifteen subjects' brain activity was recorded with fMRI as they guessed what categories groups of words belong to.

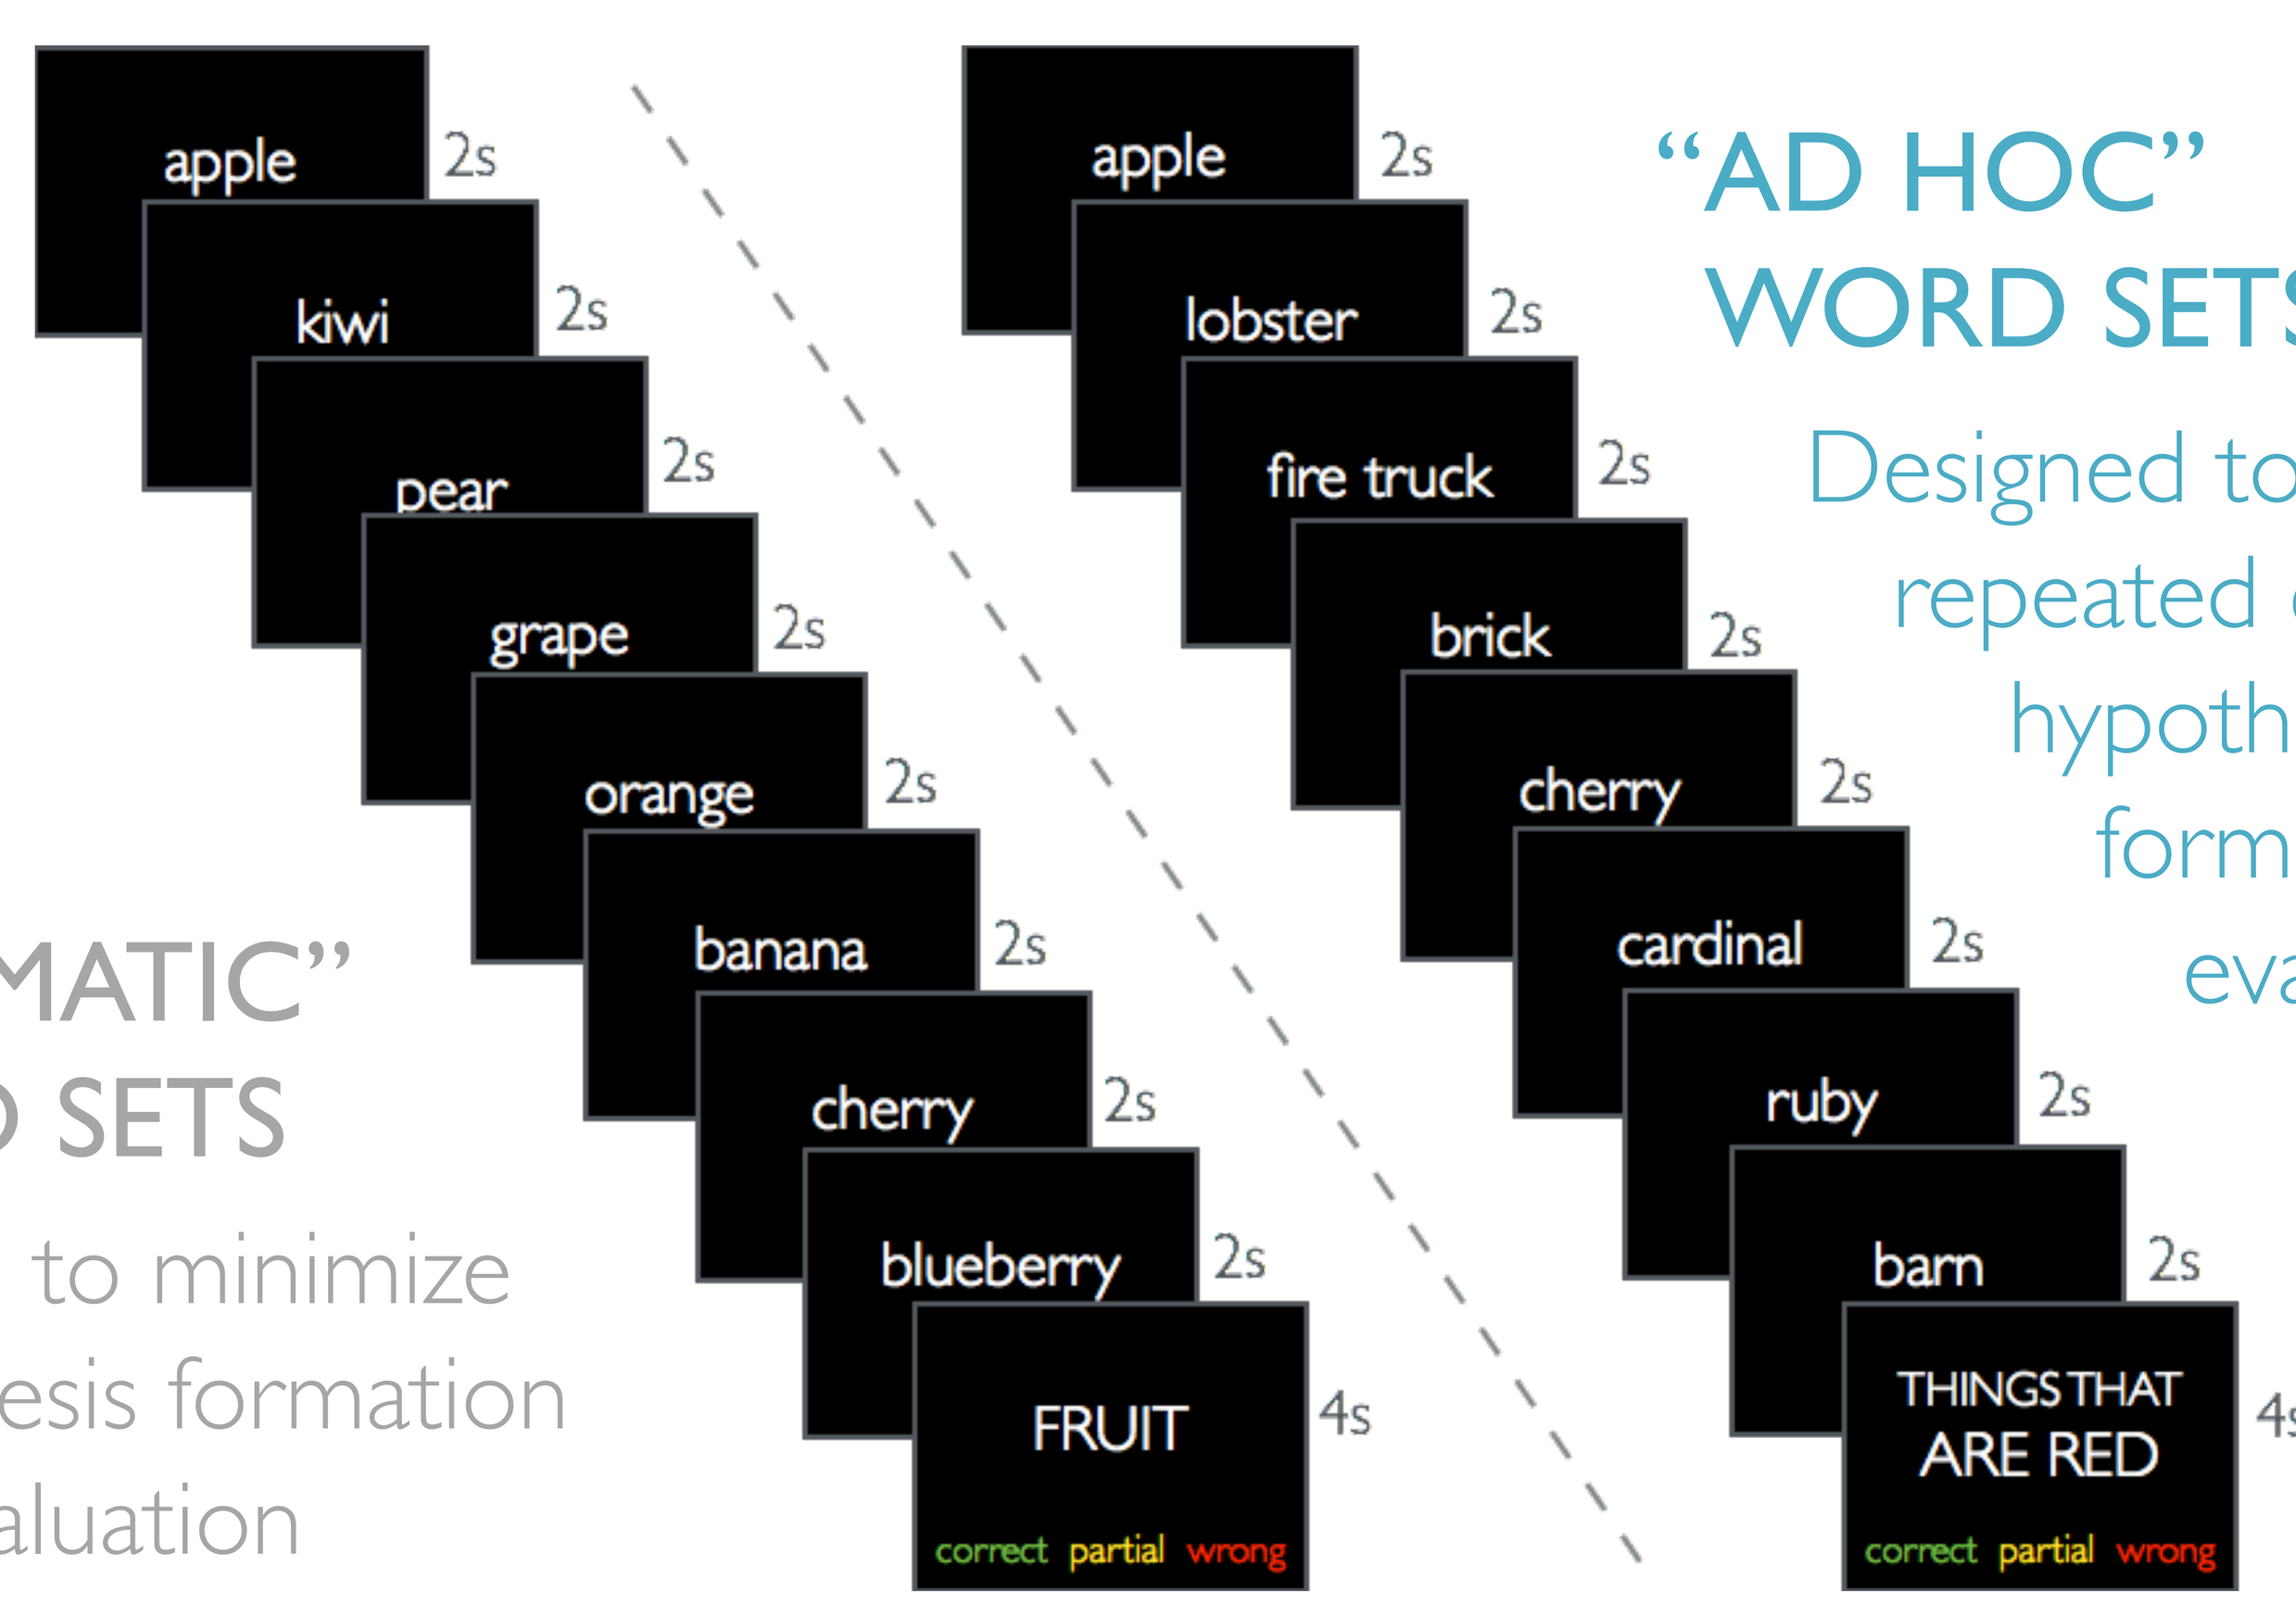
apple
2s
kiwi
pear
grape
orange
banana
cherry
blueberry
FRUIT
4s
correct partial wrong
apple
lobster
fire truck
brick
cherry
cardinal
ruby
barn
THINGS THAT ARE RED

"AD HOC" WORD SETS
Designed to elicit repeated cycles of hypothesis formation & evaluation

"AUTOMATIC" WORD SETS
Designed to minimize hypothesis formation & evaluation

Expectancy estimation

A different group of subjects were given partial word lists and generated items that fit in each list. Their responses were compiled to estimate expectancies of the words in the word sets.

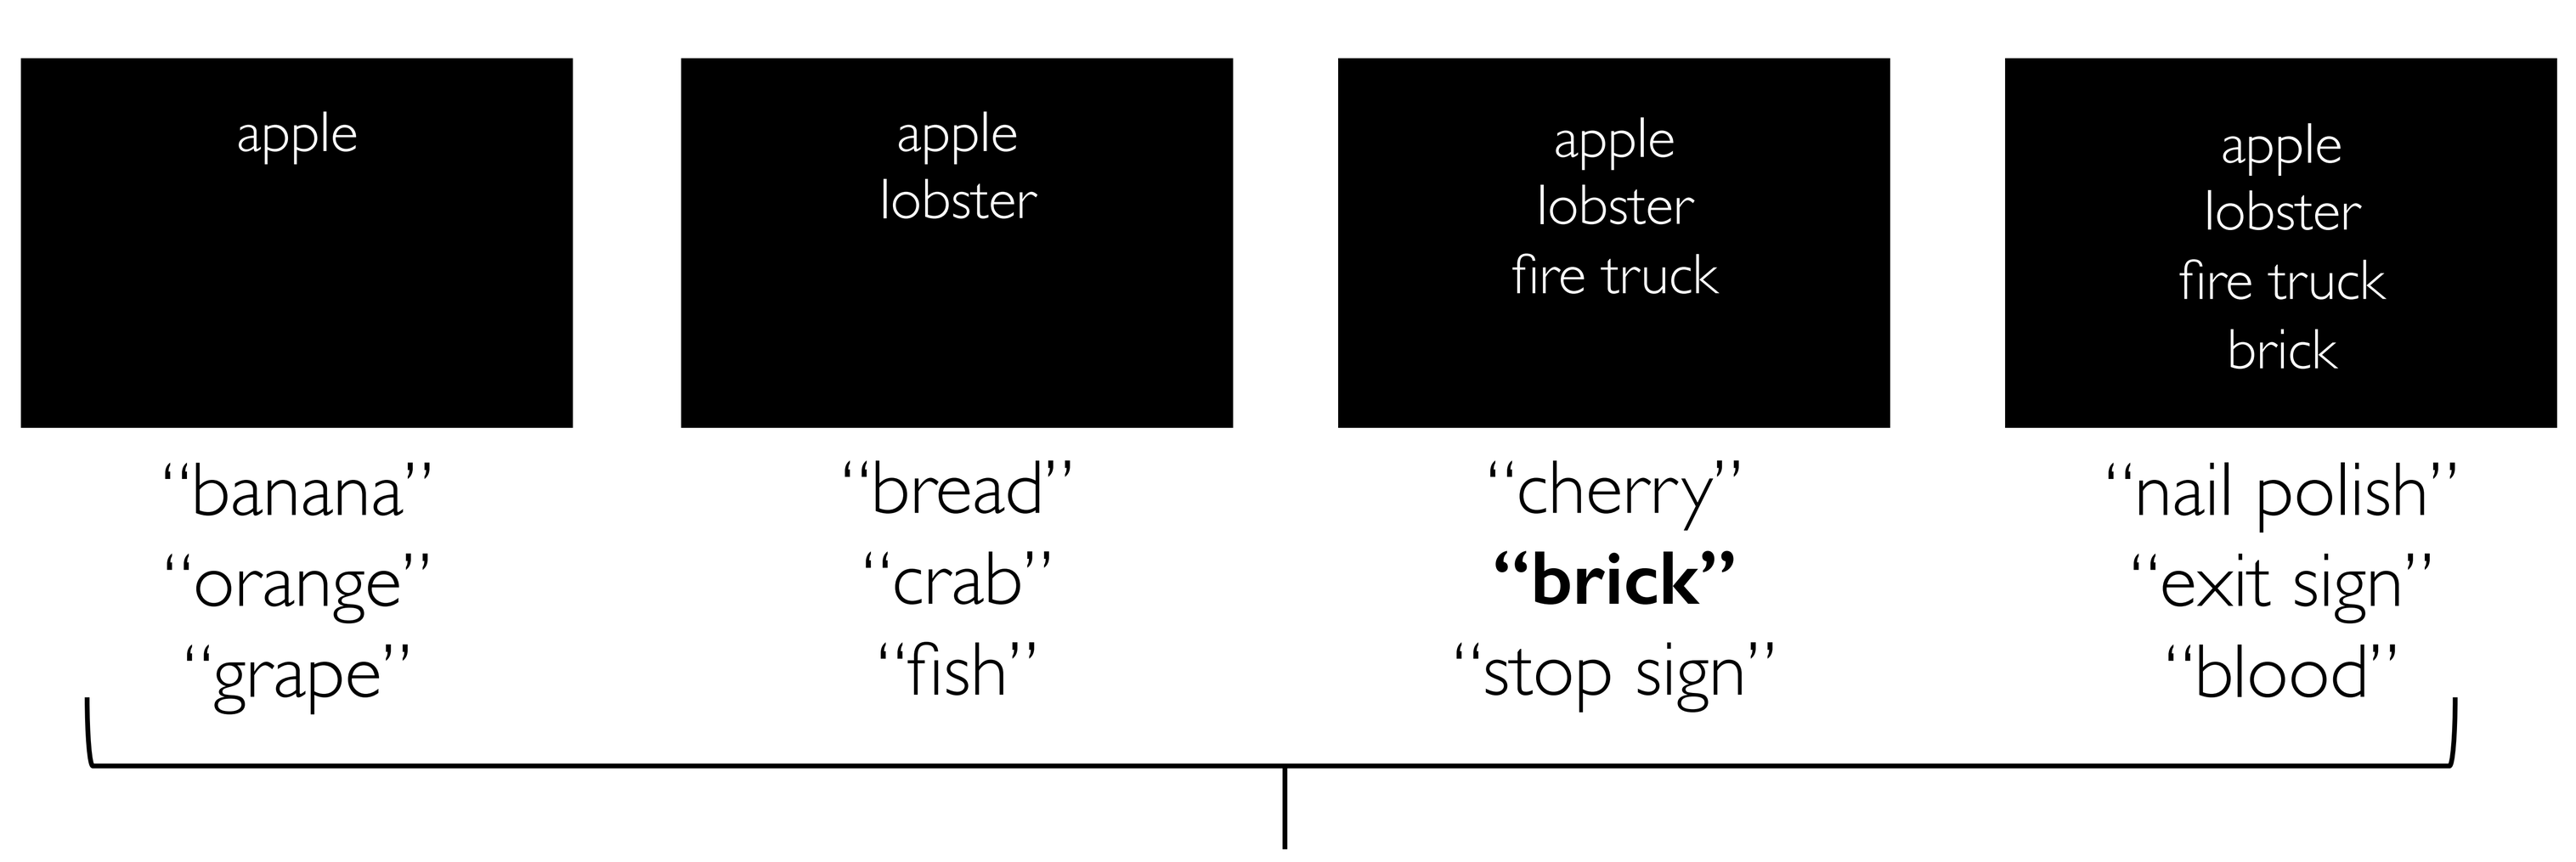
apple
"banana"
"orange"
"grape"
apple
lobster
"bread"
"crab"
"fish"
apple
lobster
fire truck
"cherry"
"brick"
"stop sign"
apple
lobster
fire truck
brick
"nail polish"
"exit sign"
"blood"

expectancy = P(word | previous words)

Expectancy results

Ad hoc word sets violate expectations more than automatic word sets.

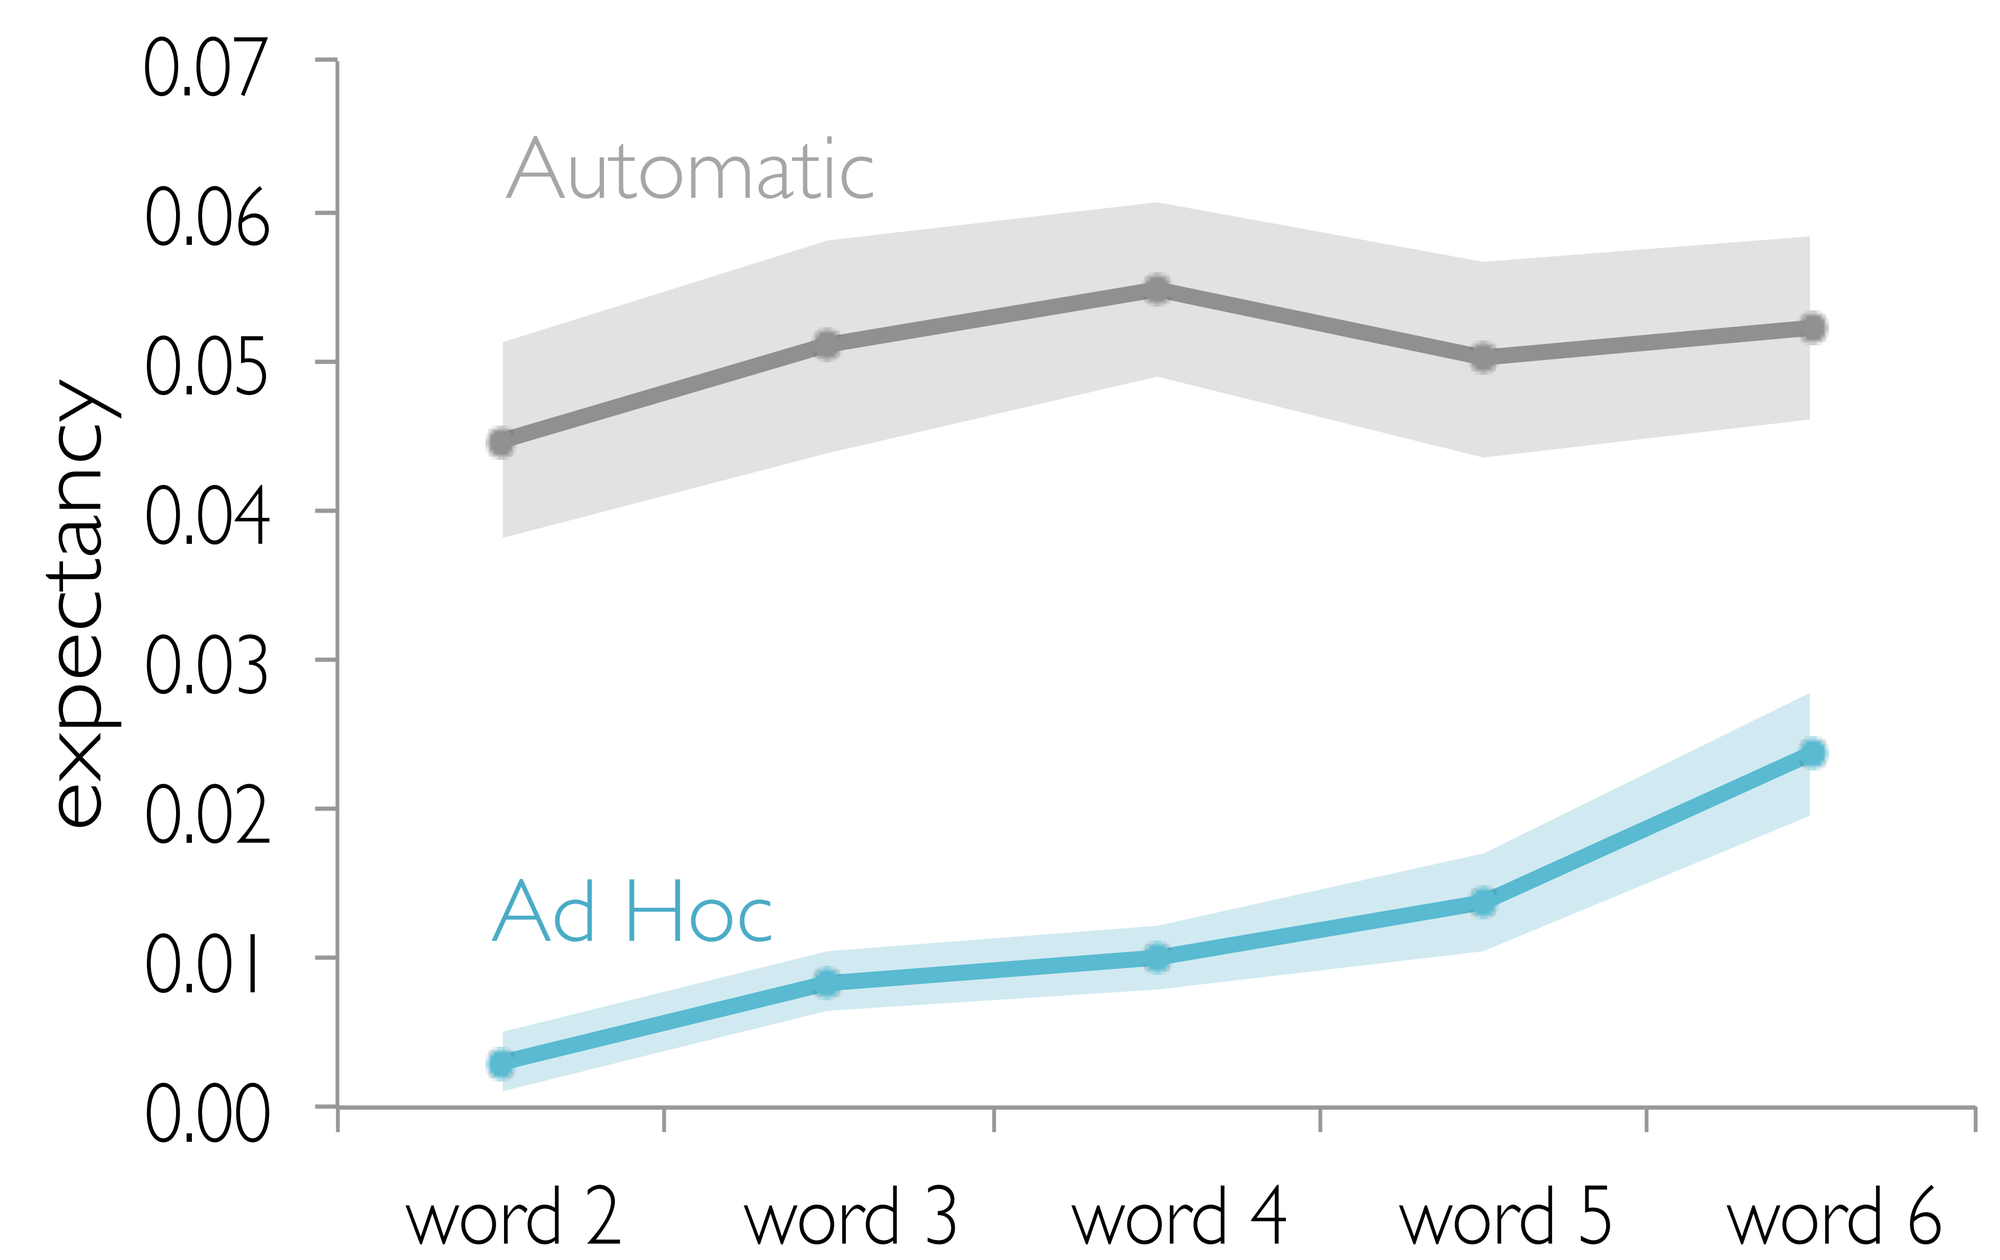
0.07
0.06
0.05
0.04
0.03
0.02
0.01
0.00
expectancy
Automatic
Ad Hoc
word 2
word 3
word 4
word 5
word 6

Word sets with unexpected words take longer to solve.

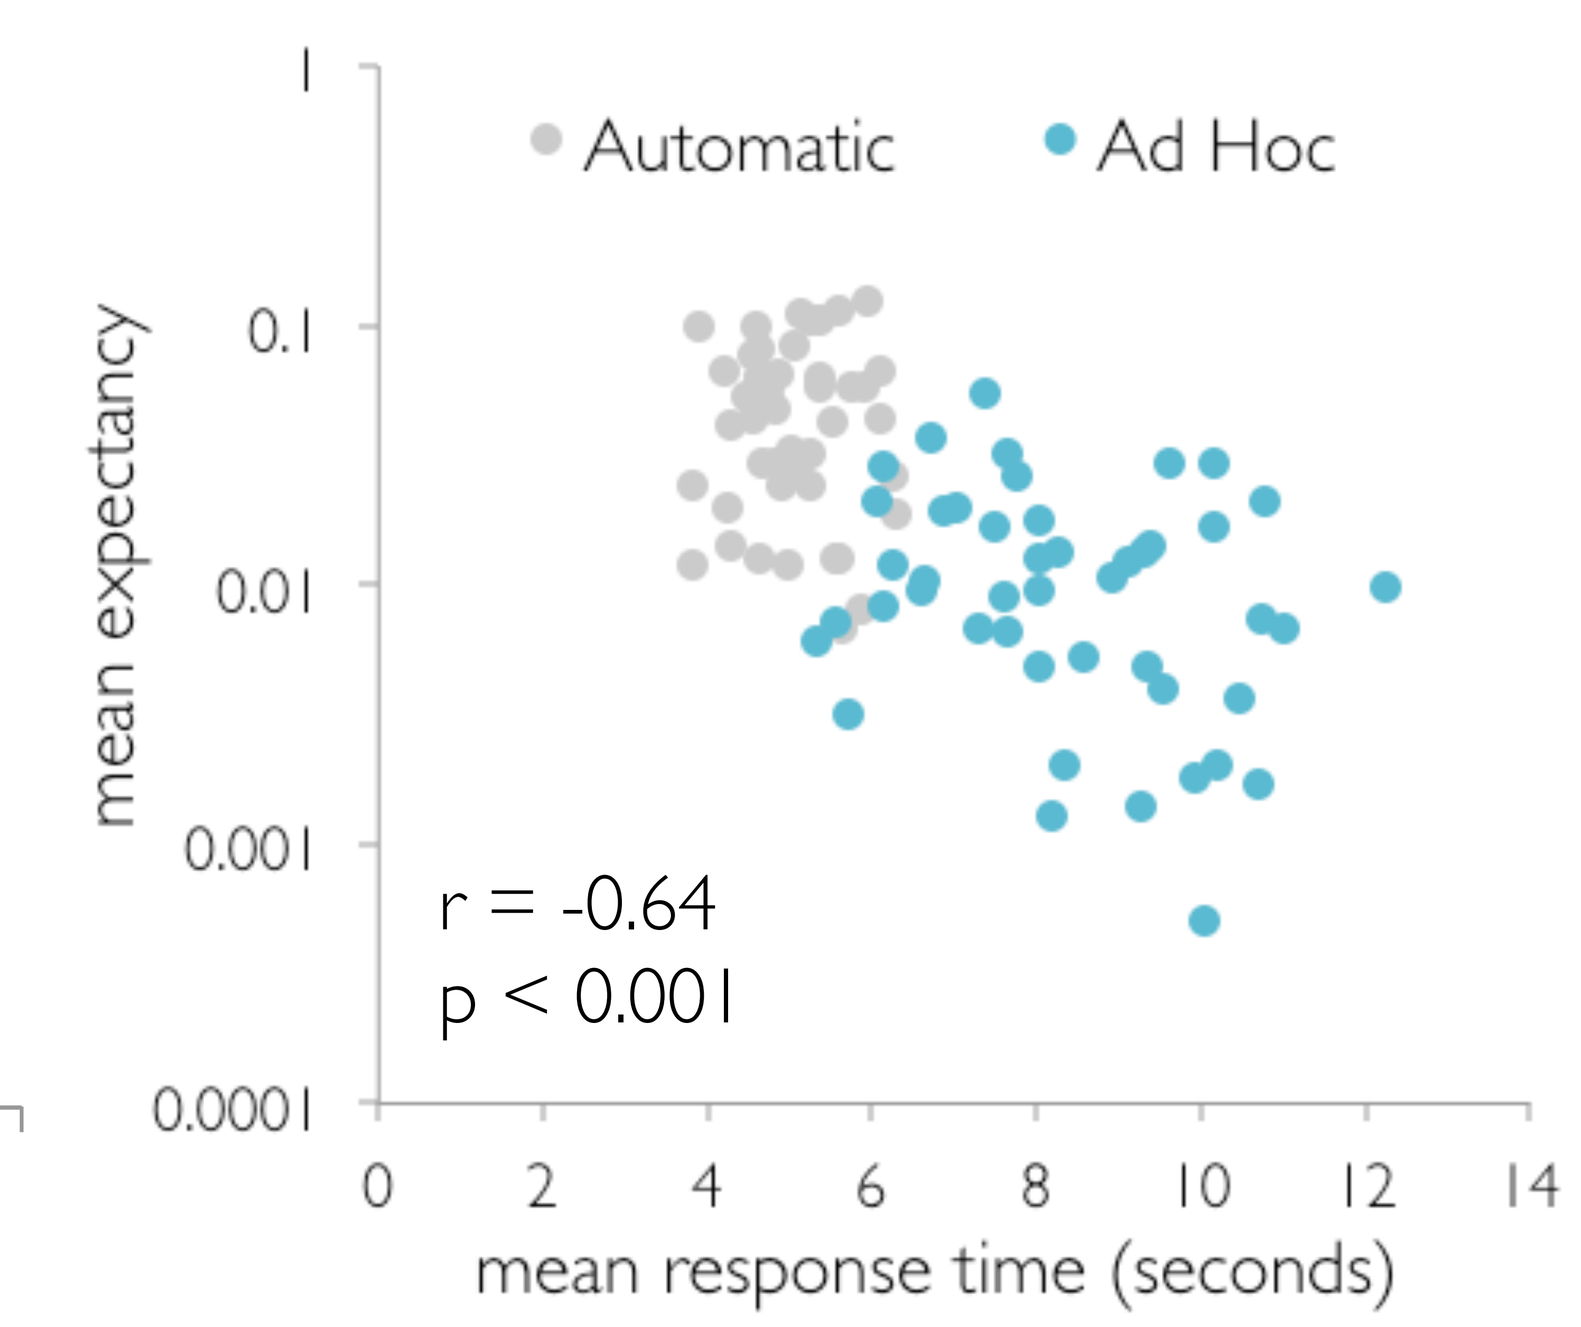
1
0.1
0.01
0.001
0.0001
mean expectancy
Automatic
Ad Hoc
r = -0.64
p < 0.001
0
2
4
6
8
10
12
14
mean response time (seconds)

Expectation violation in the brain

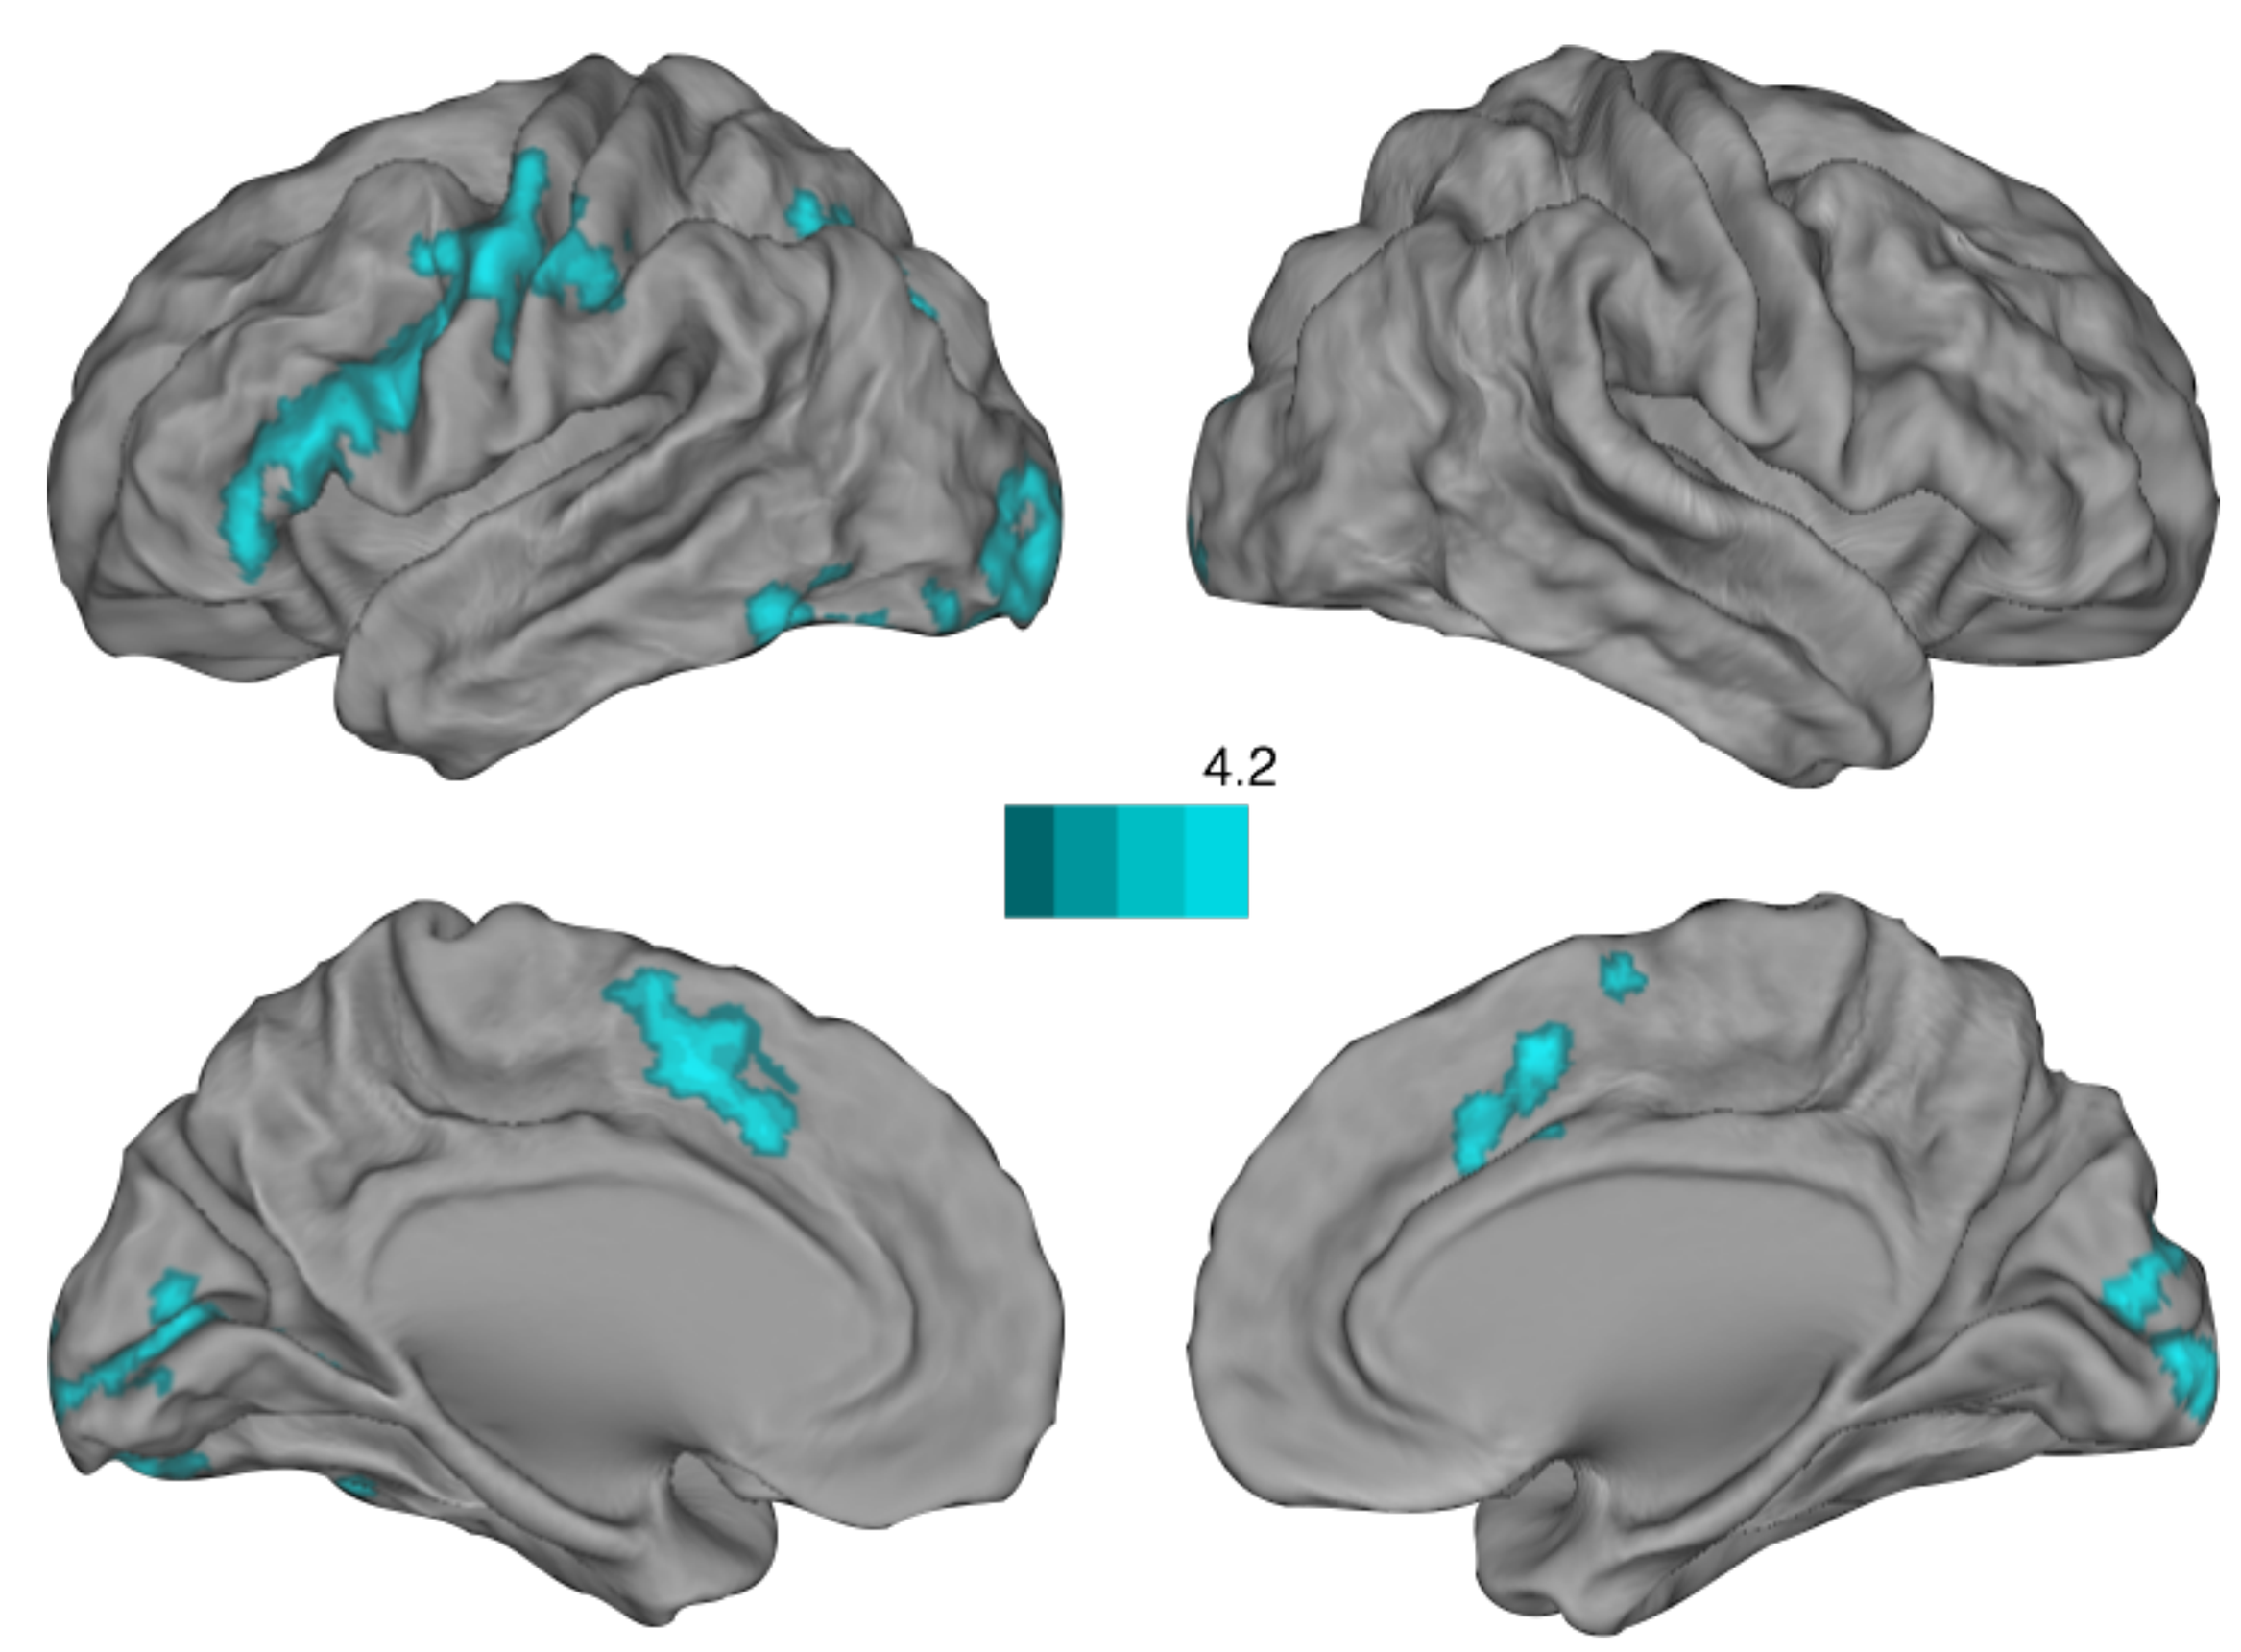
4.2

Brain areas in blue are active when a word violates expectations.
We propose that these areas – especially the left vlPFC and dmPFC – support the search and evaluation of new explanations.

Bayesian model & parameter estimation

$$\widehat{P}(H|word) = \frac{\widehat{P}(H) * \widehat{P}(word|H)}{\widehat{P}(H) * \widehat{P}(word|H) + \widehat{P}(\sim H) * \widehat{P}(word|\sim H)}$$

Exp. 1: present first word, subjects list possible categories

Exp. 2: present category, subjects list category members

word frequency

$$H = correct\ category$$

Bayesian results

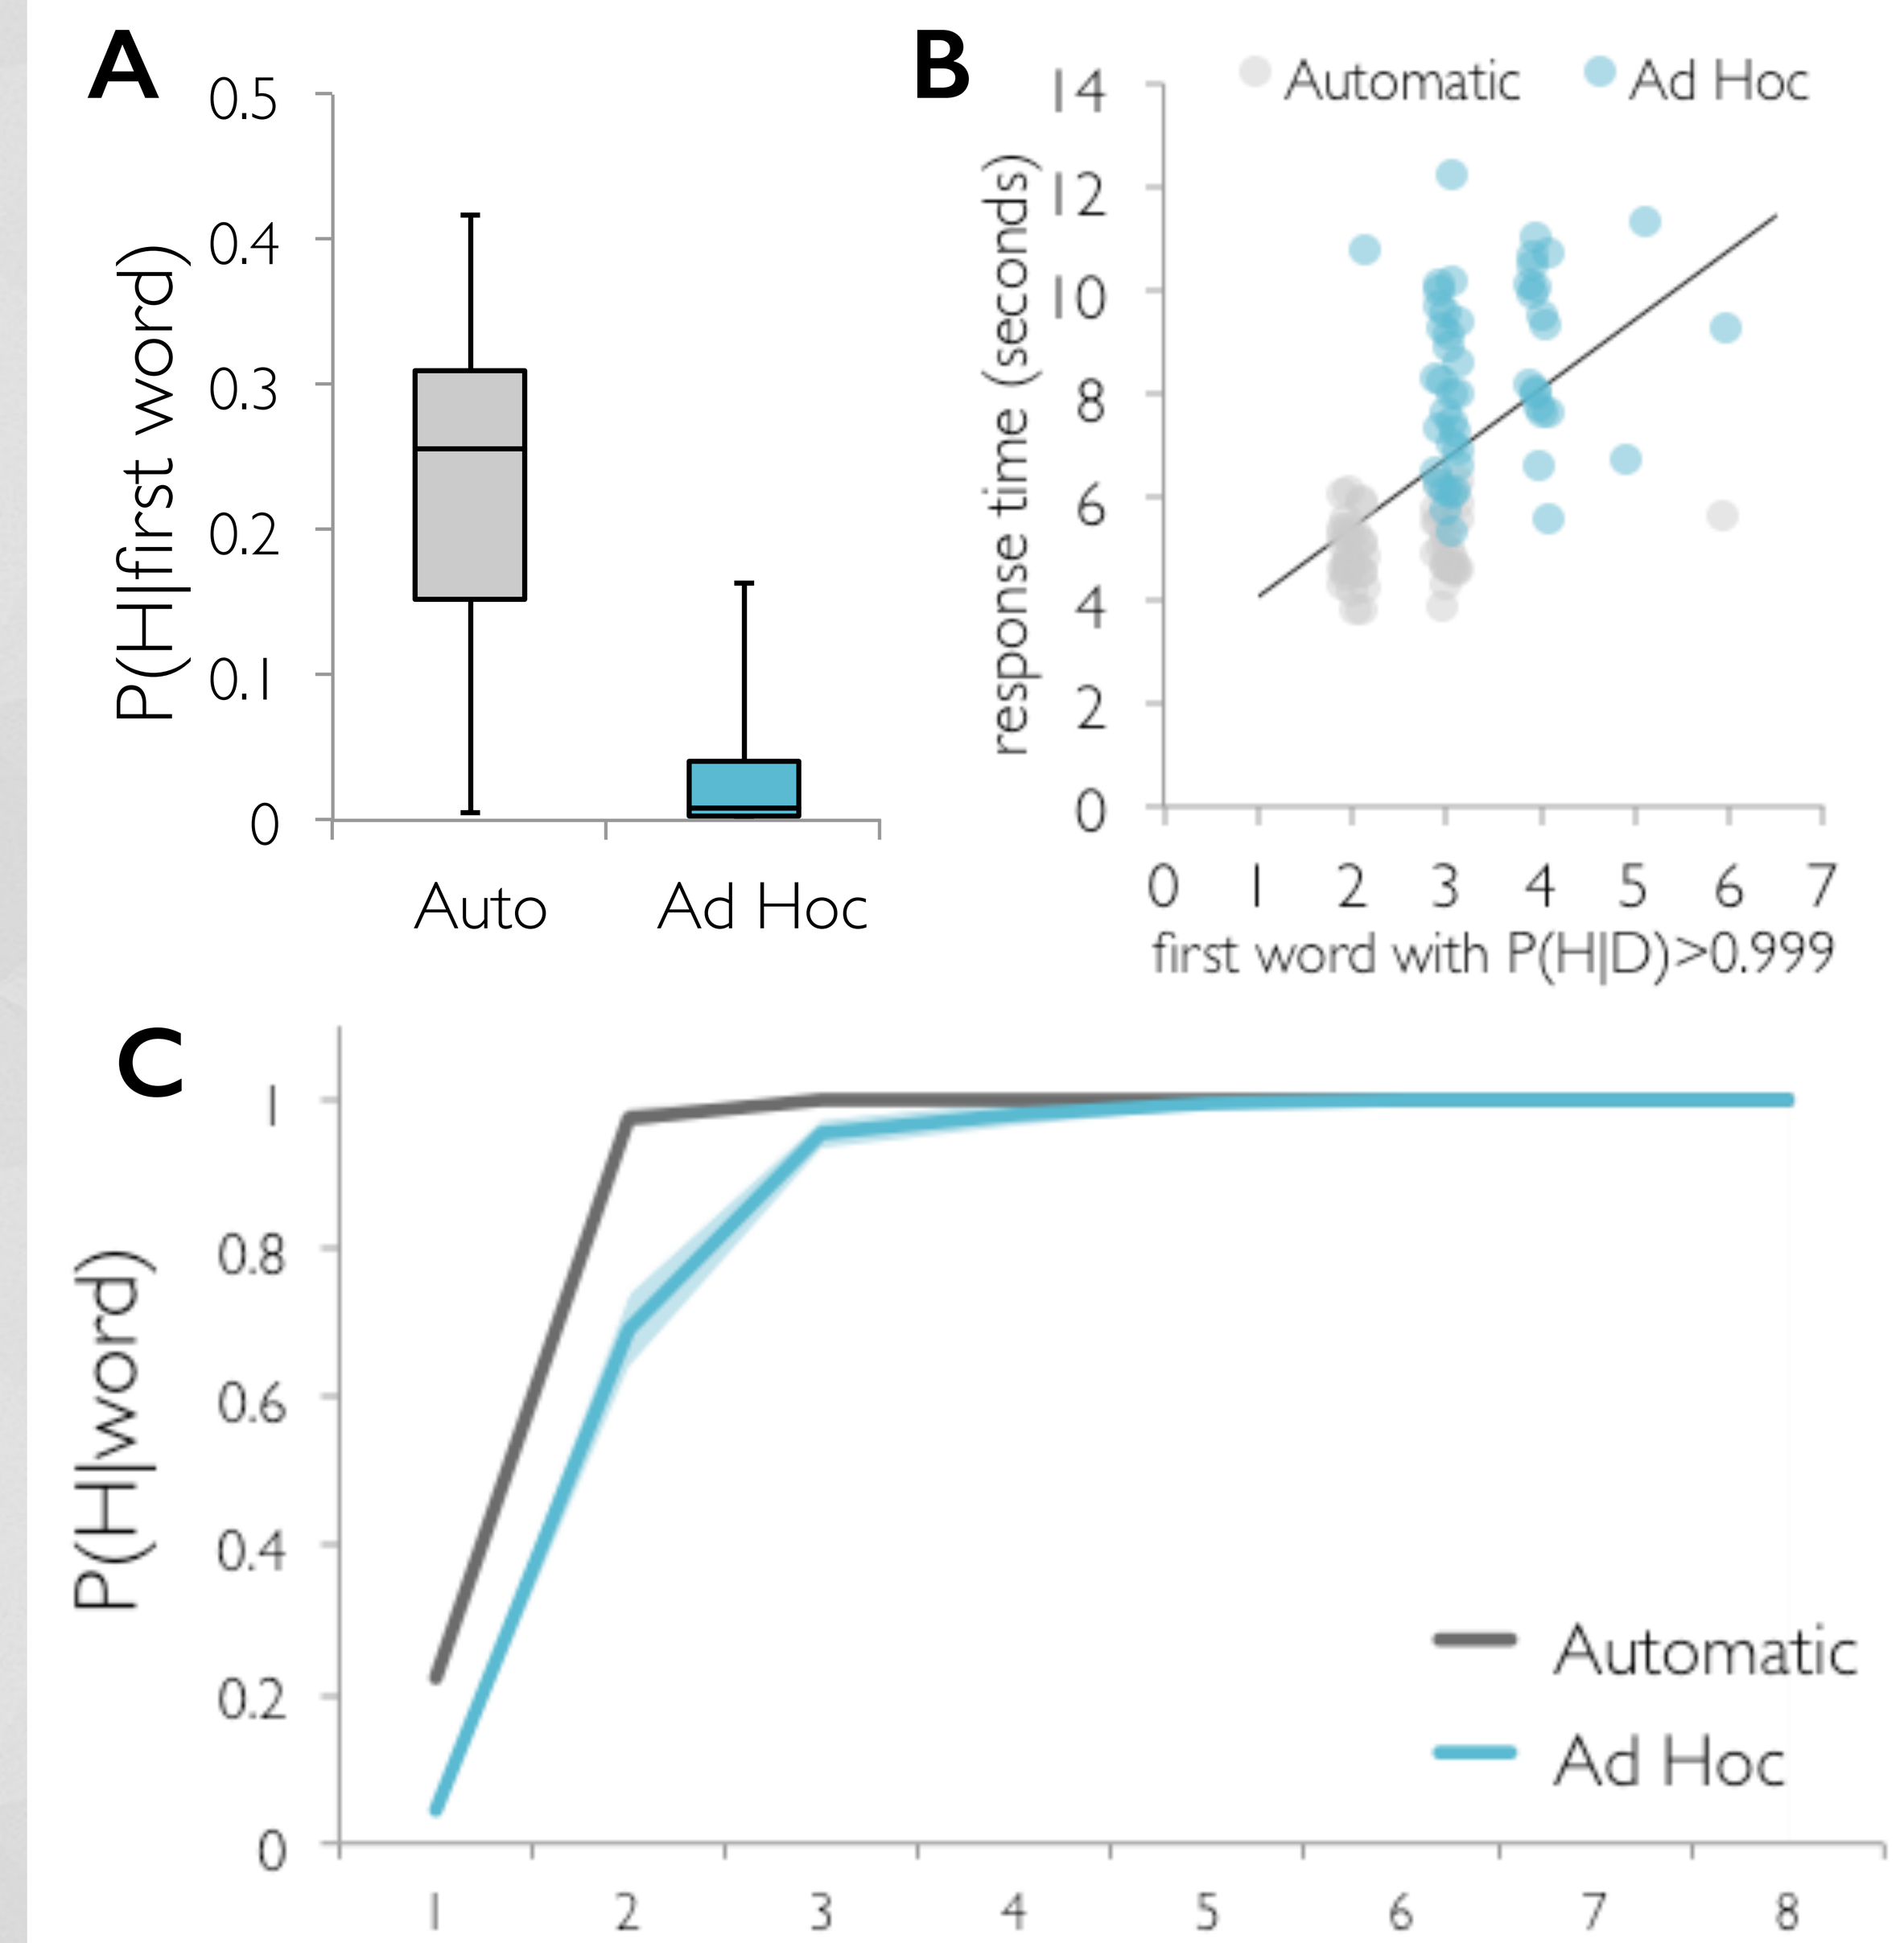
A
0.5
0.4
0.3
0.2
0.1
0
P(H|first word)
Auto
Ad Hoc
B
14
12
10
8
6
4
2
0
response time (seconds)
Automatic
Ad Hoc
0 1 2 3 4 5 6 7
first word with P(H|D)>0.999
C
1
0.8
0.6
0.4
0.2
0
P(H|word)
Automatic
Ad Hoc
1 2 3 4 5 6 7 8

A Subjects' initial ad hoc category hypotheses tend to be incorrect.
B Hypothesis strength is related to response time.
C Hypothesis strength increases over the course of the trial, and tends to increase faster in automatic trials than in ad hoc trials.

Discussion

Observations that are inconsistent with current beliefs violate expectations and necessitate the search for a new hypothesis. The left vlPFC and dmPFC are sensitive to expectation violations, suggesting they may support this search.

We plan to refine and expand the Bayesian model and use its predictions to isolate brain regions that are associated with subcomponents of hypothesis making.

References

1. Coltheart, M. (2010). The neuropsychology of delusions. *Ann. N.Y. Acad. Sci., 1191*, 16-26.
2. Gettys, C., & Fisher, S. (1979). Hypothesis plausibility and hypothesis generation. *Organizational Behavior and Human Performance, 24*, 93-110.
3. Landmann, C., et al. (2006). Dynamics of prefrontal and cingulate activity during a reward-based logical deduction task. *Cerebral Cortex, 17*, 749-759.

This research was funded by the Institute for Collaborative Biotechnologies under grant W911NF-09-D-0001.

marinsek@psych.ucsb.edu